Building towards a computational infrastructure to aid in the interpretation of Bicycle® toxin conjugate response profiles

Sean Santos[1], David Galbraith[2], Dan Rozelle[2], Judy Chuk[1], Phil Jeffrey[3], Steve Blakemore[1], Tara Gelb[1]

[1]Bicycle Therapeutics, 4 Hartwell Place, Lexington, MA, USA; [2]Rancho Bio Sciences, San Diego, CA, USA; [3]Bicycle Therapeutics, Babraham Research Campus, Cambridge, UK

ABSTRACT

- Our clinical stage Bicycle toxin conjugates® (BTCs) (BT1718, BT5528, BT8009) consist of a tumor antigen targeting bicyclic peptide linked to a microtubule binding agent (MTBA) payload via a molecular spacer and cleavable linker.
 - Early signs of clinical activity have been reported for BT1718 and BT5528 (the two most advanced BTCs) in the ongoing Phase I/II trials.
- The aim of this study was to build an integrated computational infrastructure to understand factors likely to influence response to BTCs™, including tumor antigen expression and MTBA BTC payload sensitivity.
 - We identified 181 genes whose transcript expression was associated with sensitivity to MTBAs.
 - Low ABCB1 expression was associated with MTBA sensitivity, and MTBAs were more commonly approved in cancer indications with relatively low ABCB1 expression.
 - Transcript expression of BTC-tumor antigens (e.g. MT1-MMP (BT1718), EphA2 (BT5528) & Nectin-4(BT8009)) was integrated into our computational pipeline.
- We anticipate use of a multiplex approach, which includes quantification of BTC tumor antigen and ABCB1 expression to help elucidate BTC response profiles.

INTRODUCTION

- Bicycles are fully synthetic constrained peptides with antibody-like affinities and target selectivity that readily penetrate tumor tissue, have relatively short half-lives, and can be chemically linked to a toxin to generate BTCs.

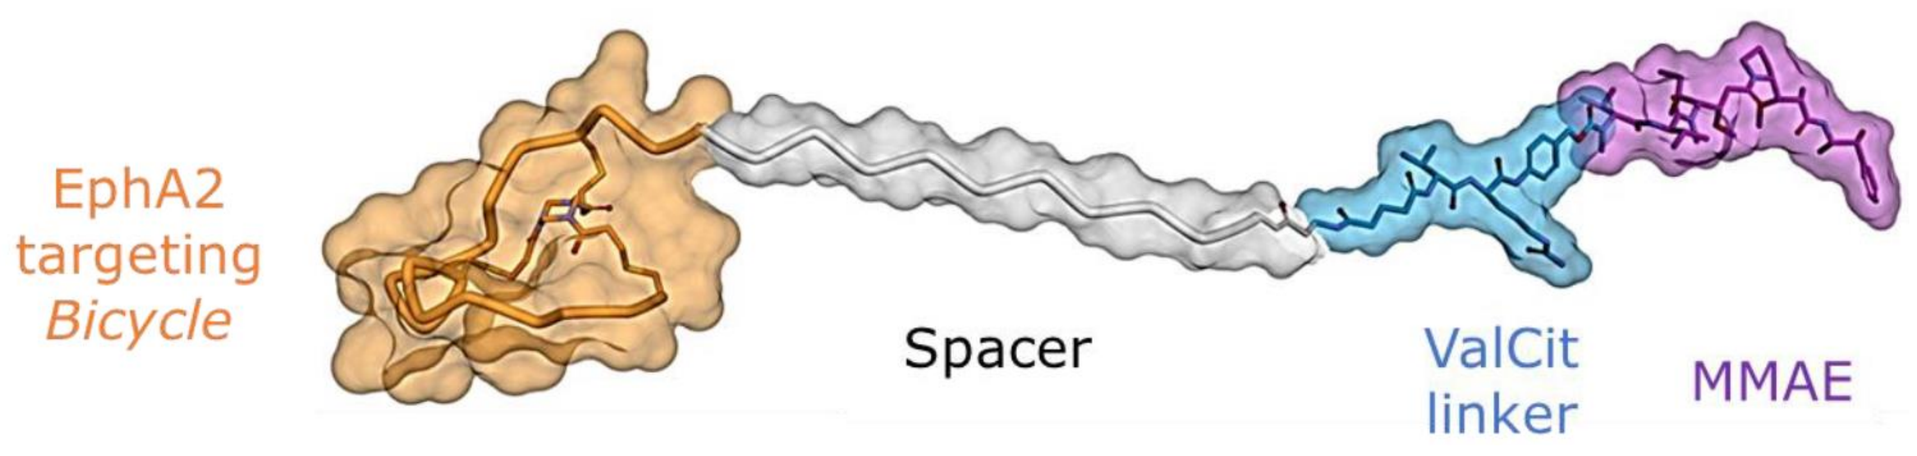
EphA2 targeting *Bicycle*
Spacer
ValCit linker
MMAE

Figure 1: BT5528, a BTC consists of an EphA2 tumor antigen targeting bicyclic peptide linked to MMAE (an MTBA) via a molecular spacer and cleavable (ValCit) linker.

- ABCB1, which is an ATP-dependent efflux pump (aliases: P-glycoprotein 1 (PGP); multidrug resistance protein 1 (MDR1)), can pump MTBAs and other drugs out of the cell and has been implicated in resistance to MTBAs.

METHODS

- Regression modeling was performed on the Genentech cell line screening initiative (gCSI) dataset [1] from PharmacoDB (accessed on 17-Nov-2020) [2], which contains both RNA-seq and efficacy readouts (dose-response curves) to identify genes potentially involved in MTBA sensitivity.
- Area above the dose-response curve (AAC) was used as a measure of efficacy and potency.
- In the linear regression model of gene-drug sensitivity, genes that are significantly associated with a sensitivity measure have a non-zero slope, where the AAC changes with gene expression. An FDR corrected p-value threshold of < 0.01 was used as a cutoff.
- ABCB1 transcript expression across cancer indications was quantified using The Cancer Genome Atlas (TCGA) dataset [3] (accessed on 8-Jan-2021), MTBA clinical use was measured using TCGA and *GlobalData* (accessed on 3-Mar-2021), and MTBA clinical approval status was determined using *Medscape* [5] (accessed on 9-Mar-2021).
 - *GlobalData* was used to estimate the clinical use of MTBAs in cancer indications using the following: *1) filter for tubulin inhibitors as mechanism, 2) remove drug combinations, 3) filter for marketed drugs, 4) count # of times drug was developed/indication (eliminate duplicates if drug was developed in multiple regions).*
 - For MTBAs in *Medscape* (paclitaxel, docetaxel, taxol, oraxol, docefrez, Taxotere), approval status was determined by cancer indication and followed a hierarchy of 1) approved indication, 2) orphan designations, or 3) off-label use. MTBA status was annotated as the highest order designation and if no MTBAs were used the indication was listed as "not approved." (e.g. HNSC was listed as off-label for paclitaxel but was approved for docetaxel → HNSC = approved for taxanes).

RESULTS

Figure 2: Identification of 181 genes predicted to influence MTBA sensitivity.

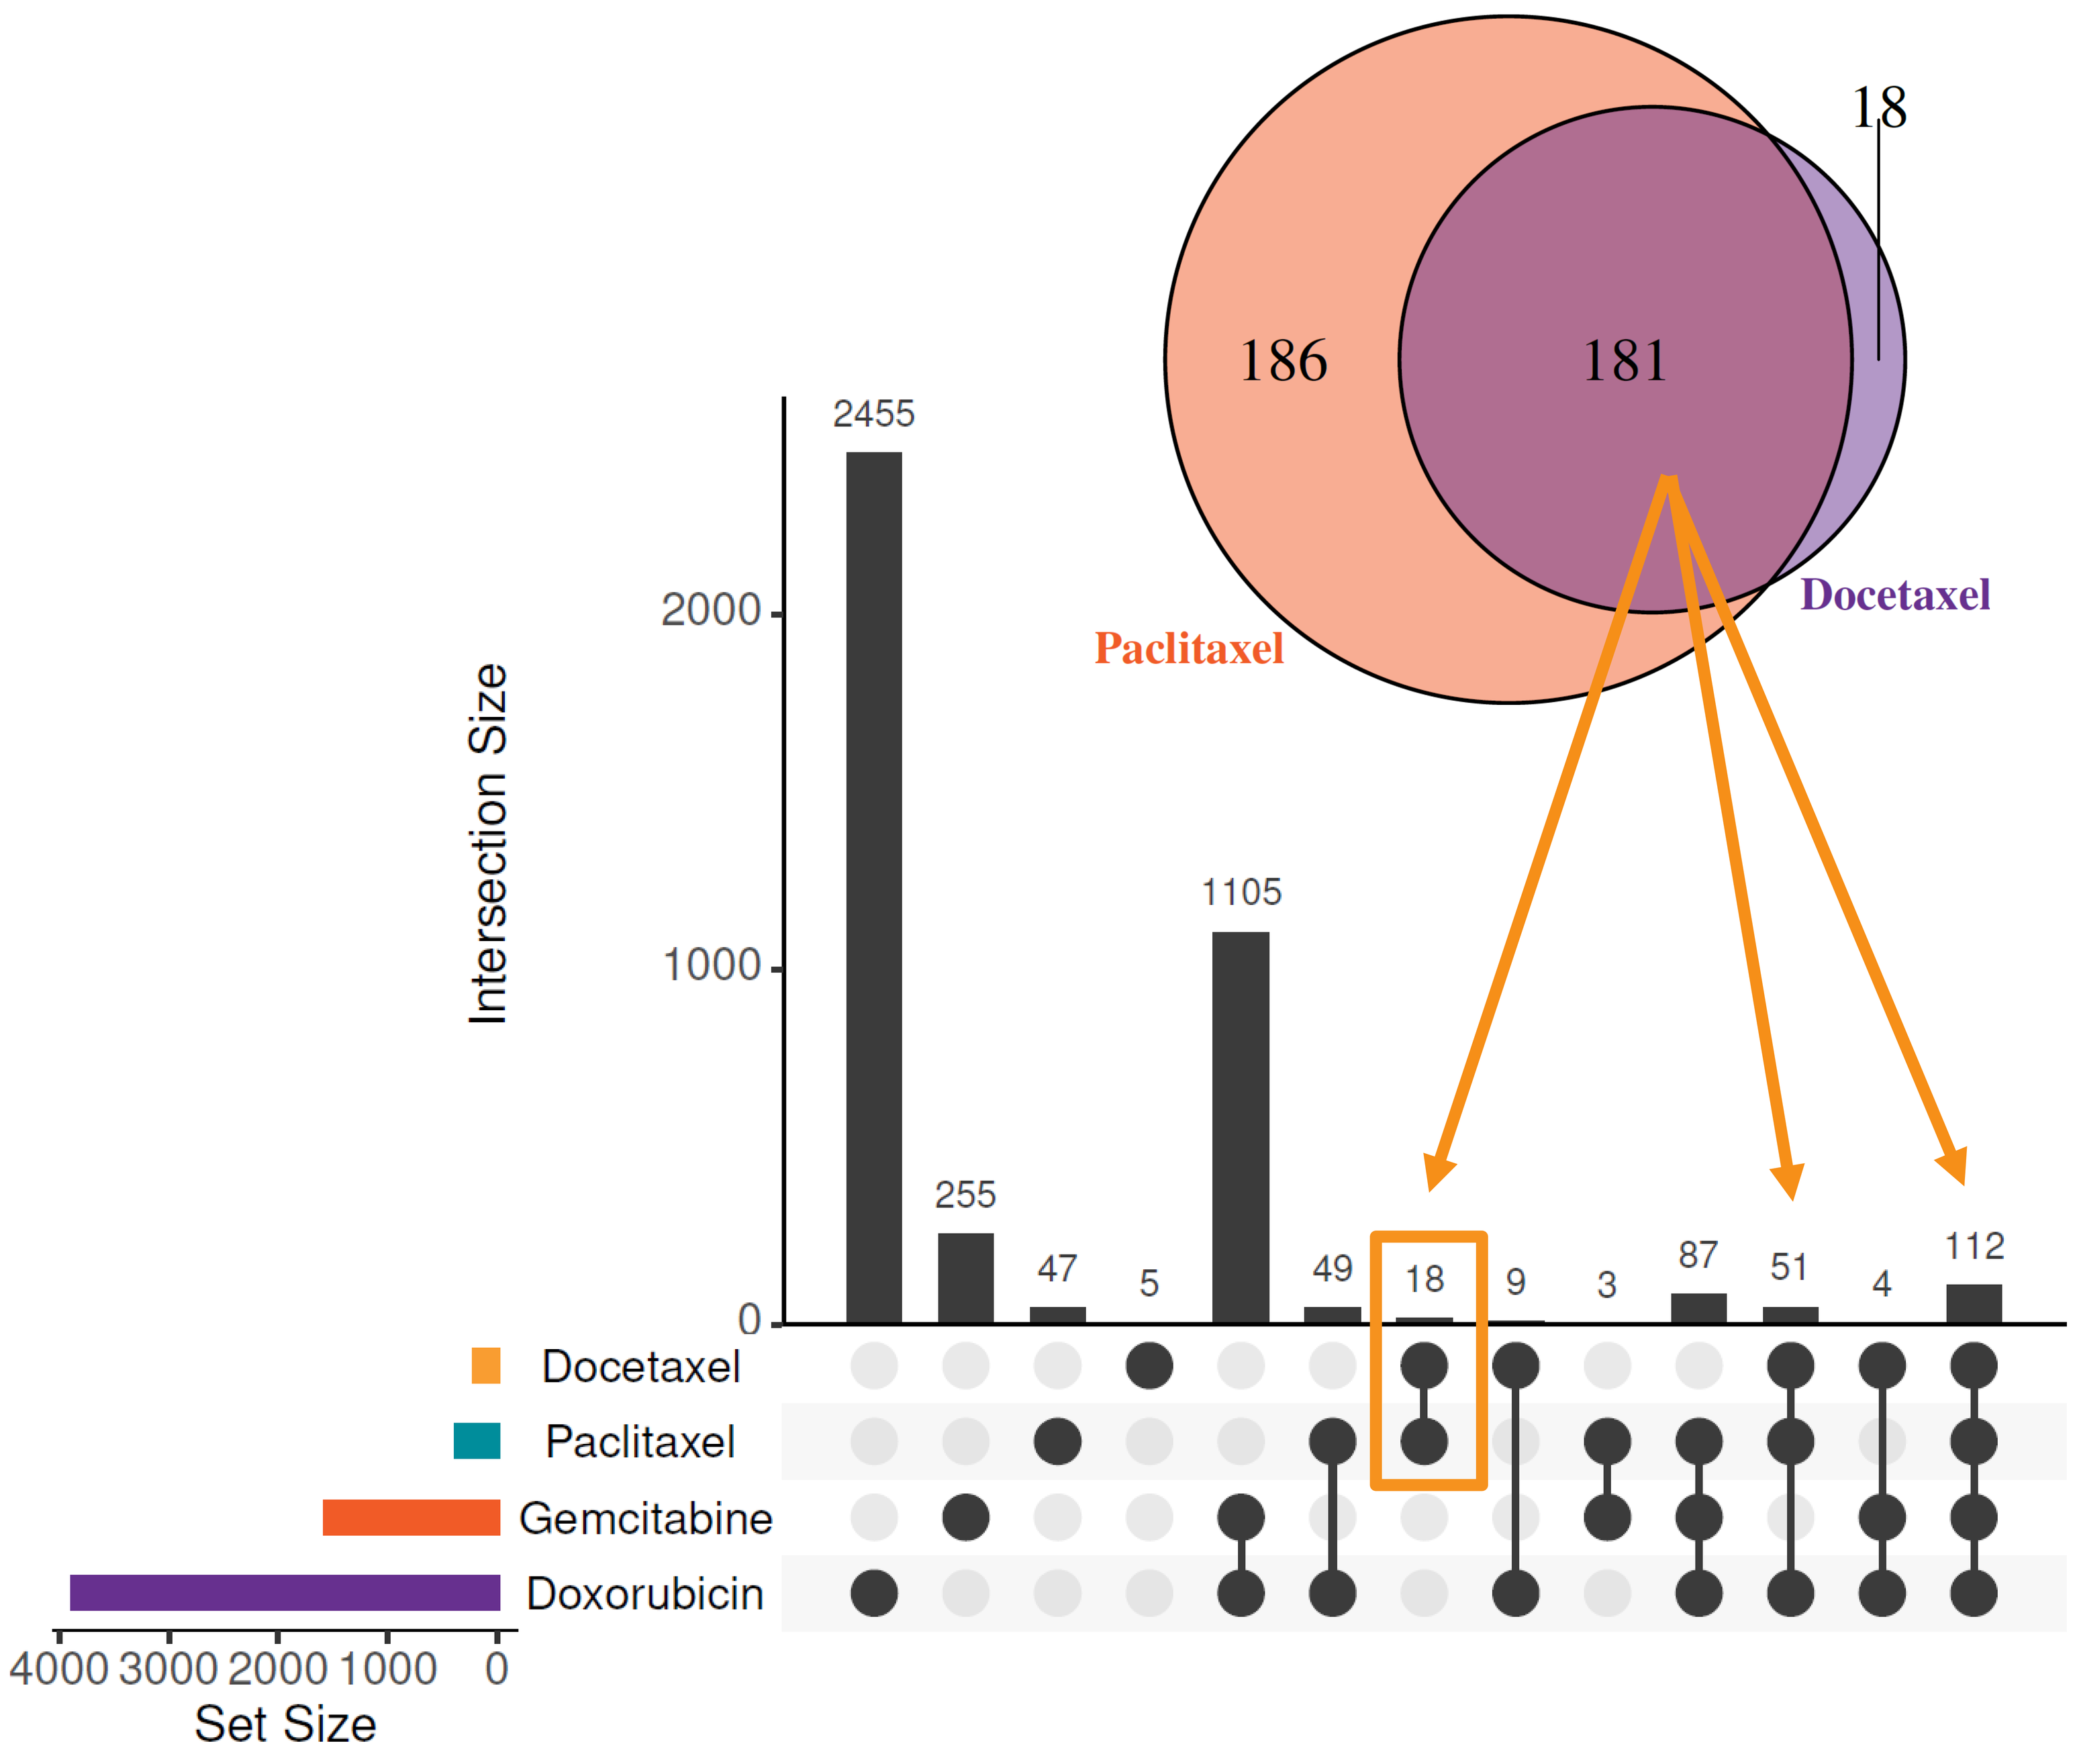
18
186
181
Paclitaxel
Docetaxel
Intersection Size
2455
255
47
5
1105
49
18
9
3
87
51
4
112
Docetaxel
Paclitaxel
Gemcitabine
Doxorubicin
4000 3000 2000 1000 0
Set Size

Analysis of the gCSI dataset in PharmacoDB led to the identification of 181 genes associated with both paclitaxel and docetaxel (two MTBAs) sensitivity (false discovery rate (FDR) < 0.01). 18 of the 181 genes, including ABCB1, were associated with paclitaxel and docetaxel sensitivity (as defined by AAC) but not doxorubicin or gemcitabine (non-MTBA cytotoxic agents) sensitivity, suggesting these 18 genes may be specifically implicated in the mechanism of action of MTBAs.

Figure 3: Reduced ABCB1 expression is strongly associated with MTBA sensitivity.

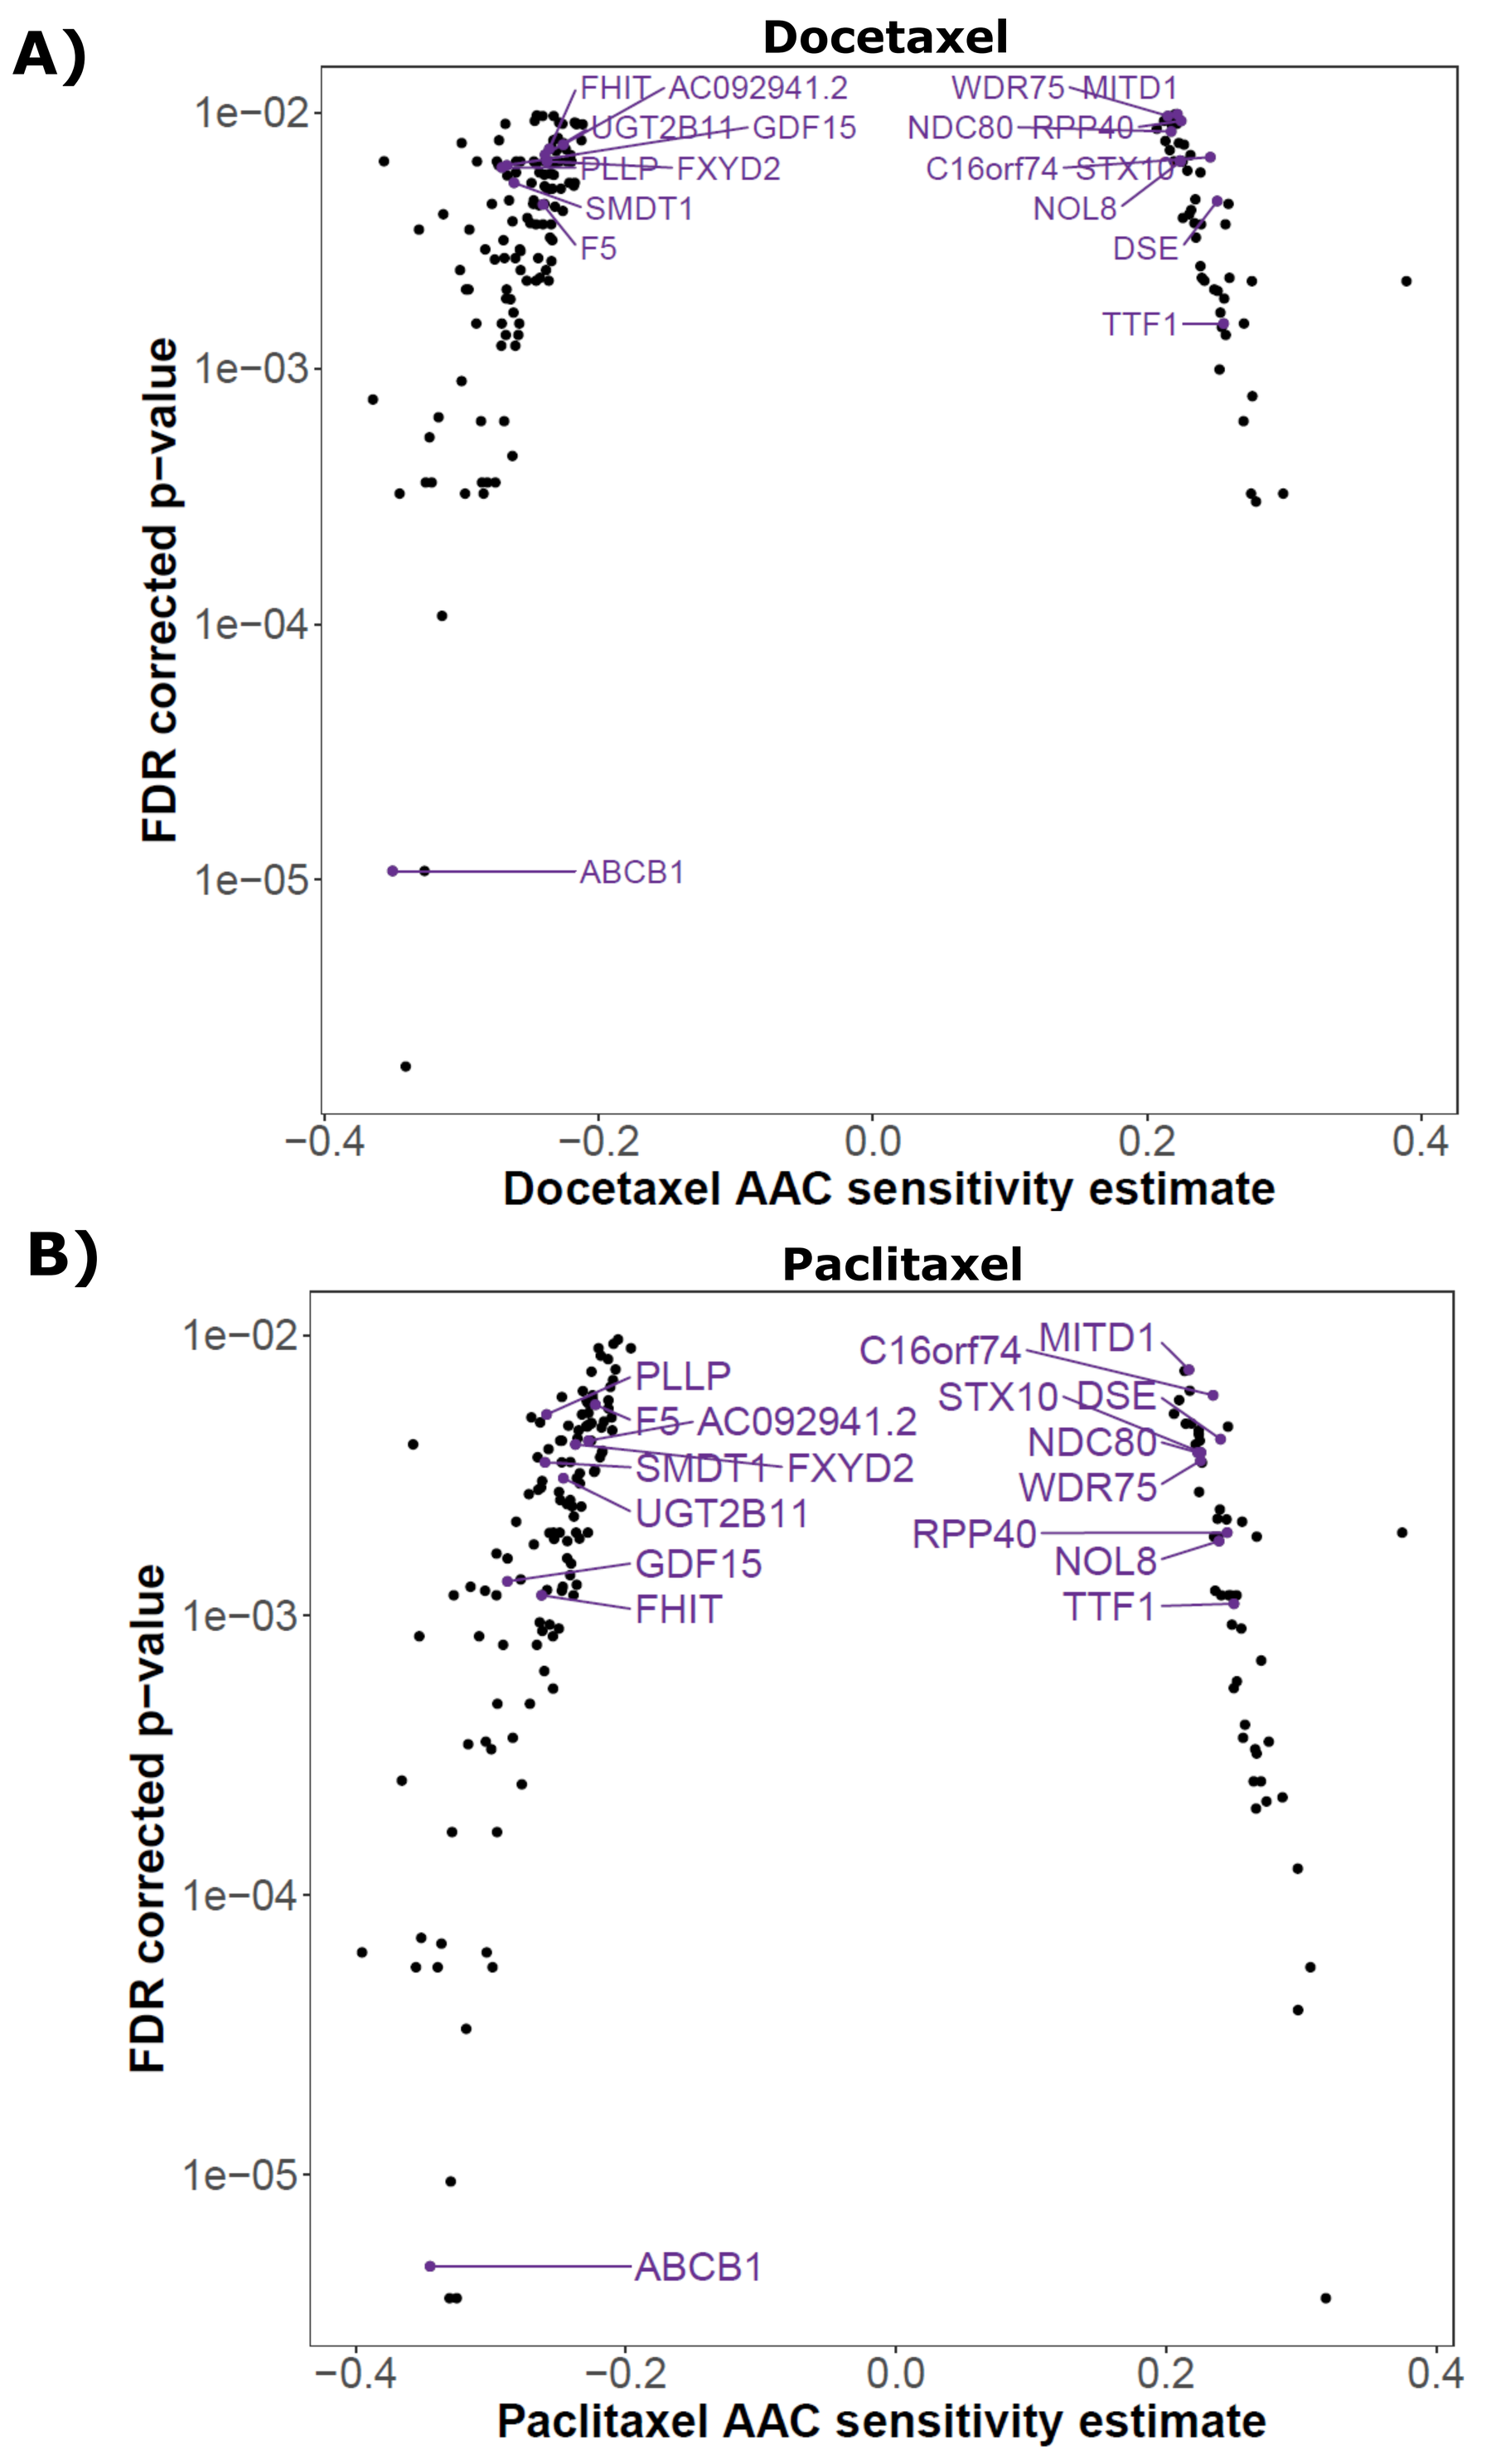
A)
Docetaxel
FHIT AC092941.2
UGT2B11 GDF15
PLLP FXYD2
SMDT1
F5
WDR75 MITD1
NDC80 RPP40
C16orf74 STX10
NOL8
DSE
TTF1
ABCB1
FDR corrected p-value
Docetaxel AAC sensitivity estimate
B)
Paclitaxel
PLLP
F5 AC092941.2
SMDT1 FXYD2
UGT2B11
GDF15
FHIT
C16orf74 MITD1
STX10 DSE
NDC80
WDR75
RPP40
NOL8
TTF1
ABCB1
FDR corrected p-value
Paclitaxel AAC sensitivity estimate

A) Docetaxel and B) paclitaxel sensitivity using AAC is plotted against FDR corrected p-value. The 18 genes that are associated with sensitivity to MTBAs, but not doxorubicin or gemcitabine are highlighted in purple (see Fig. 2).

Figure 4: MTBA agents are more commonly approved in cancer indications with lower relative ABCB1 expression

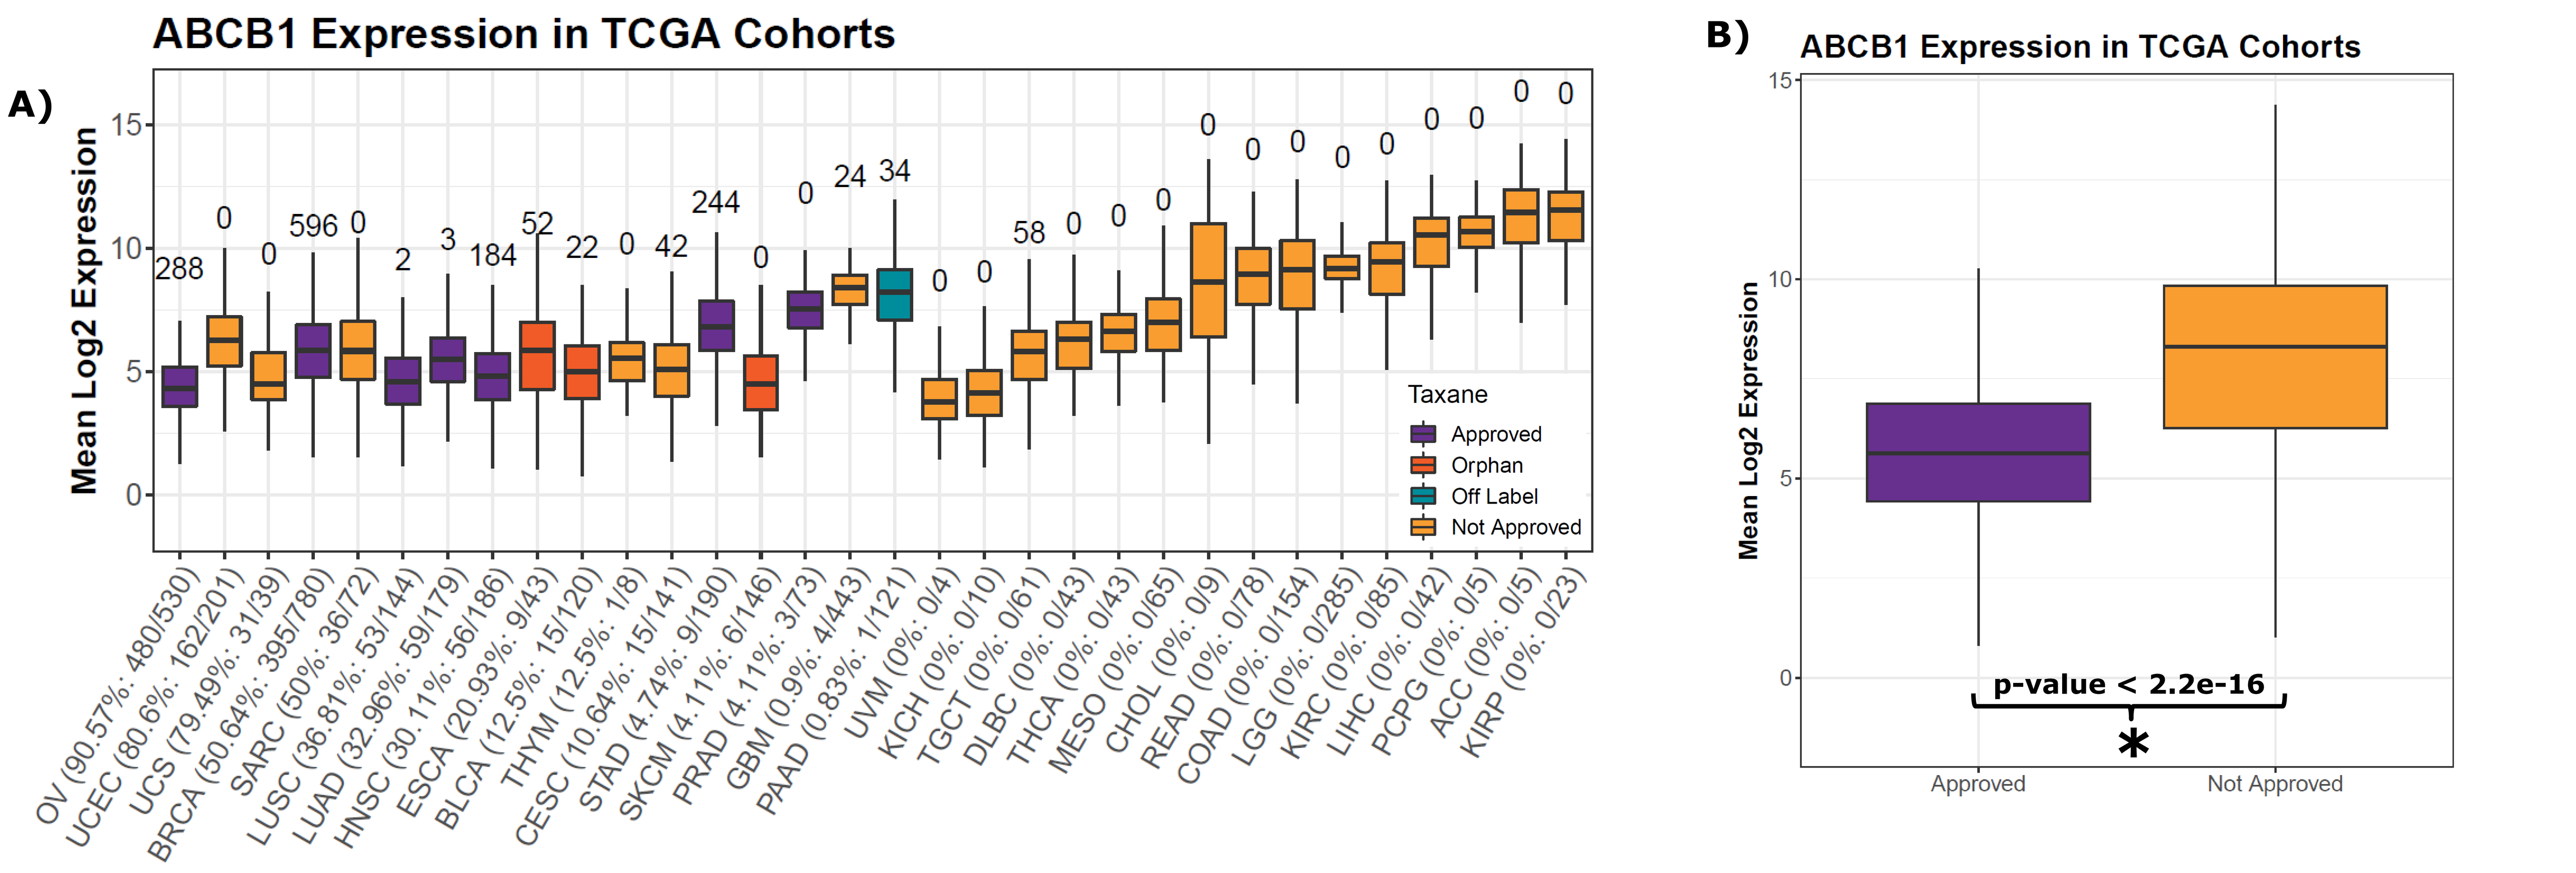
A)
ABCB1 Expression in TCGA Cohorts
Mean Log2 Expression
Taxane
Approved
Orphan
Off Label
Not Approved
OV (90.57%: 480/530)
UCEC (80.6%: 162/201)
UCS (79.49%: 31/39)
BRCA (50.64%: 395/780)
SARC (50%: 36/72)
LUSC (36.81%: 53/144)
LUAD (32.96%: 59/179)
HNSC (30.11%: 56/186)
ESCA (20.93%: 9/43)
BLCA (12.5%: 15/120)
THYM (12.5%: 1/8)
CESC (10.64%: 15/141)
STAD (4.74%: 9/190)
SKCM (4.11%: 6/146)
PRAD (4.11%: 3/73)
GBM (0.9%: 4/443)
PAAD (0.83%: 1/121)
UVM (0%: 0/4)
KICH (0%: 0/10)
TGCT (0%: 0/61)
DLBC (0%: 0/43)
THCA (0%: 0/43)
MESO (0%: 0/65)
CHOL (0%: 0/9)
READ (0%: 0/78)
COAD (0%: 0/154)
LGG (0%: 0/285)
KIRC (0%: 0/85)
LIHC (0%: 0/42)
PCPG (0%: 0/5)
ACC (0%: 0/5)
KIRP (0%: 0/23)
B)
ABCB1 Expression in TCGA Cohorts
Mean Log2 Expression
p-value < 2.2e-16
*
Approved
Not Approved

A) Indications are ordered based on the percentage of patients (see x-axis label) treated with a MTBA out of the total number of patients in that TCGA cohort. The number above each boxplot indicates the number of *GlobalData* entries (see Methods). B) ABCB1 expression was statistically significantly lower in tumor samples from TCGA for patients in MTBA approved cancer indications compared to indications where MTBAs are not approved as determined using a Wilcoxon rank sum test (*p-value < 2.2e-16).

Figure 5: BLCA has relatively low ABCB1 RNA expression and relatively high A) EphA2 and B) Nectin-4 RNA expression.

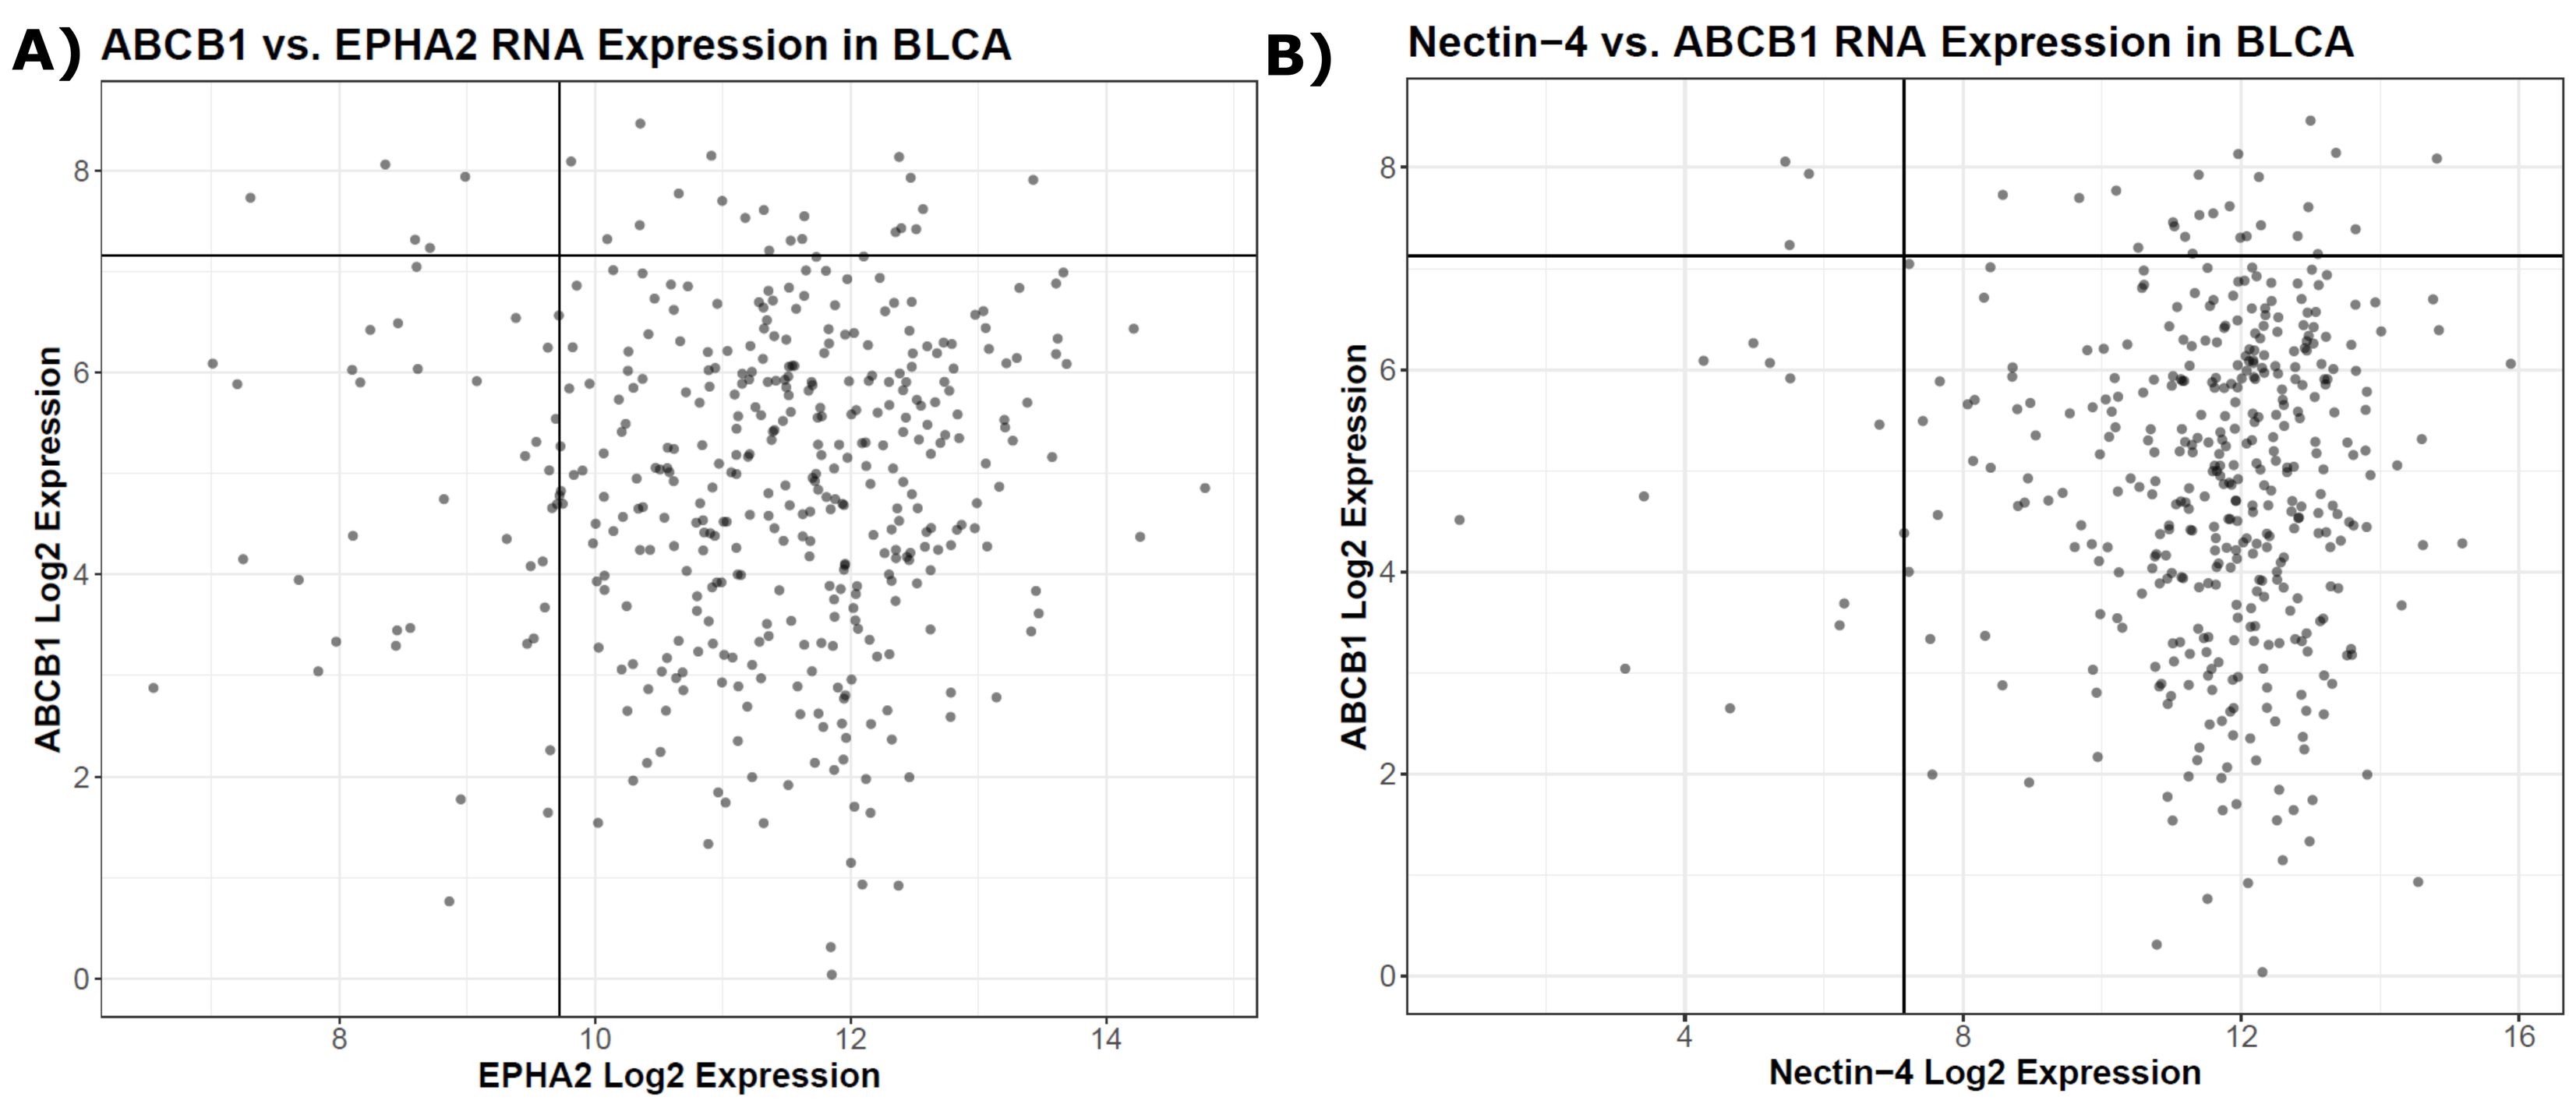
A) ABCB1 vs. EPHA2 RNA Expression in BLCA
ABCB1 Log2 Expression
EPHA2 Log2 Expression
B) Nectin−4 vs. ABCB1 RNA Expression in BLCA
ABCB1 Log2 Expression
Nectin−4 Log2 Expression

ABCB1 RNA expression in bladder urothelial carcinoma (BLCA) is plotted against **A)** EphA2 RNA expression or **B)** Nectin-4 RNA expression. Lines on the X and Y axis indicate the median expression of tumor antigen or ABCB1, respectively, across all 33 TCGA cancer indications. Expression of BTC-tumor antigens (e.g. MT1-MMP (BT1718), EphA2 (BT5528) & Nectin-4(BT8009)) vs. ABCB1 expression was integrated into our computational infrastructure for all TCGA cancer indications (not shown).

CONCLUSION/SUMMARY

- We identified genes whose transcript expression were associated with sensitivity to MTBAs.
- Our proposed computational pipeline is intended to help identify cancer indications predicted to be sensitive to MTBAs with high expression of BTC-tumor antigens
- A multifactorial approach that characterizes both target expression and MTBA sensitivity is underway to understand BTC sensitivity.
- We anticipate use of a multiplex approach that will quantify BTC tumor antigen and ABCB1 expression in addition to other potential biomarkers to help elucidate BTC response profiles.

REFERENCES

[1] Klijn et al. Nature Biotechnology. (2015) [2] Smirnov et al. Nucleic Acids Res. (2017); [3] Cerami et al. Cancer Discovery. (2012);
[4] *GlobalData* - https://www.globaldata.com/; [5] *Medscape* - https://reference.medscape.com/;
BTC clinical trials: (BT8009-100, NCT04561362); (BT5528-100,NCT04180371); (BT1718, NCT03486730).

Bicycle Therapeutics Limited
4 Hartwell Place, Lexington, MA 02421
B900, Babraham Research Campus, Cambridge, UK CB22 3AT
www.bicycletherapeutics.com